Poster 11 Predictability Of In Vivo CYP3A4 Induction By Rifampicin In Cryopreserved And Freshly Isolated Primary Human Hepatocytes

Julie Nilles and Stephanie Ruez

Department of Drug Metabolism and Pharmacokinetics, Boehringer Ingelheim Pharma GmbH & Co. KG, Birkendorfer Str. 65, 88397 Biberach an der Riss, Germany

Induction of metabolizing enzymes like cytochrome P450 (CYP) 3A4 and the simultaneous administration of CYP3A4 substrates can lead to profound decreases in the bioavailability of the concomitantly taken drugs possibly resulting in therapeutic failure. To avoid such severe drug-drug interactions, the induction potential of development drugs is tested in vitro during preclinical development. The goal was to establish a comparison of fundamental factors like hepatic cell system, read-out of induction and prediction model which can influence the predictivity of in vivo induction. Rifampicin was selected as a prototypical CYP3A4 inducer, and corresponding clinical interaction studies with midazolam as a highly sensitive CYP3A4 substrate were identified. The potency (EC_{50}) and the maximum effect (E_{max}) of induction were determined with increasing inducer concentrations in cryopreserved and freshly isolated primary human hepatocytes (PHH) after 48 hours of incubation (one single donor each). mRNA induction was determined by quantitative polymerase chain reaction (qPCR), and induction of CYP3A4 activity was detected by liquid chromatography - mass spectrometry (LC-MS/MS). The 'fold-change method', the basic kinetic model (R), and the mechanistic static model (MSM) were used as prediction models. Key findings were as follows: 1) Prediction analysis revealed exclusively true positive results for induction in cryopreserved and fresh PHH. Prediction of in vivo induction by mRNA expression in fresh PHH was significantly closer to in vivo situation compared to predictions from cryopreserved cells (GMFE R: 1.72 vs. 9.41; GMFE MSM: 2.62 vs. 89.1). The latter exclusively lead to over prediction for both induction read-outs in all prediction models. 2) mRNA expression and enzyme activity were suitable in vitro markers for in vivo induction. There was no difference in induction of mRNA and activity in freshly PHH resulting in reliable predictions by all models (GMFE: 1.72 – 2.62). In cryopreserved cells, prediction by activity assessment was closer to in vivo induction compared to prediction by mRNA expression (R 2-fold and MSM 4-fold higher accuracy). 3) All prediction models lead to true positive predictions. Induction potency in vivo (strong vs. moderate) was not always accurately predicted with 50% false predictions by the R model and 25% false predictions by the MSM revealing weaknesses in predicting induction potency. In conclusion, all tested factors lead to true positive predictions and can be used to assess in vivo CYP3A4 induction by rifampicin.

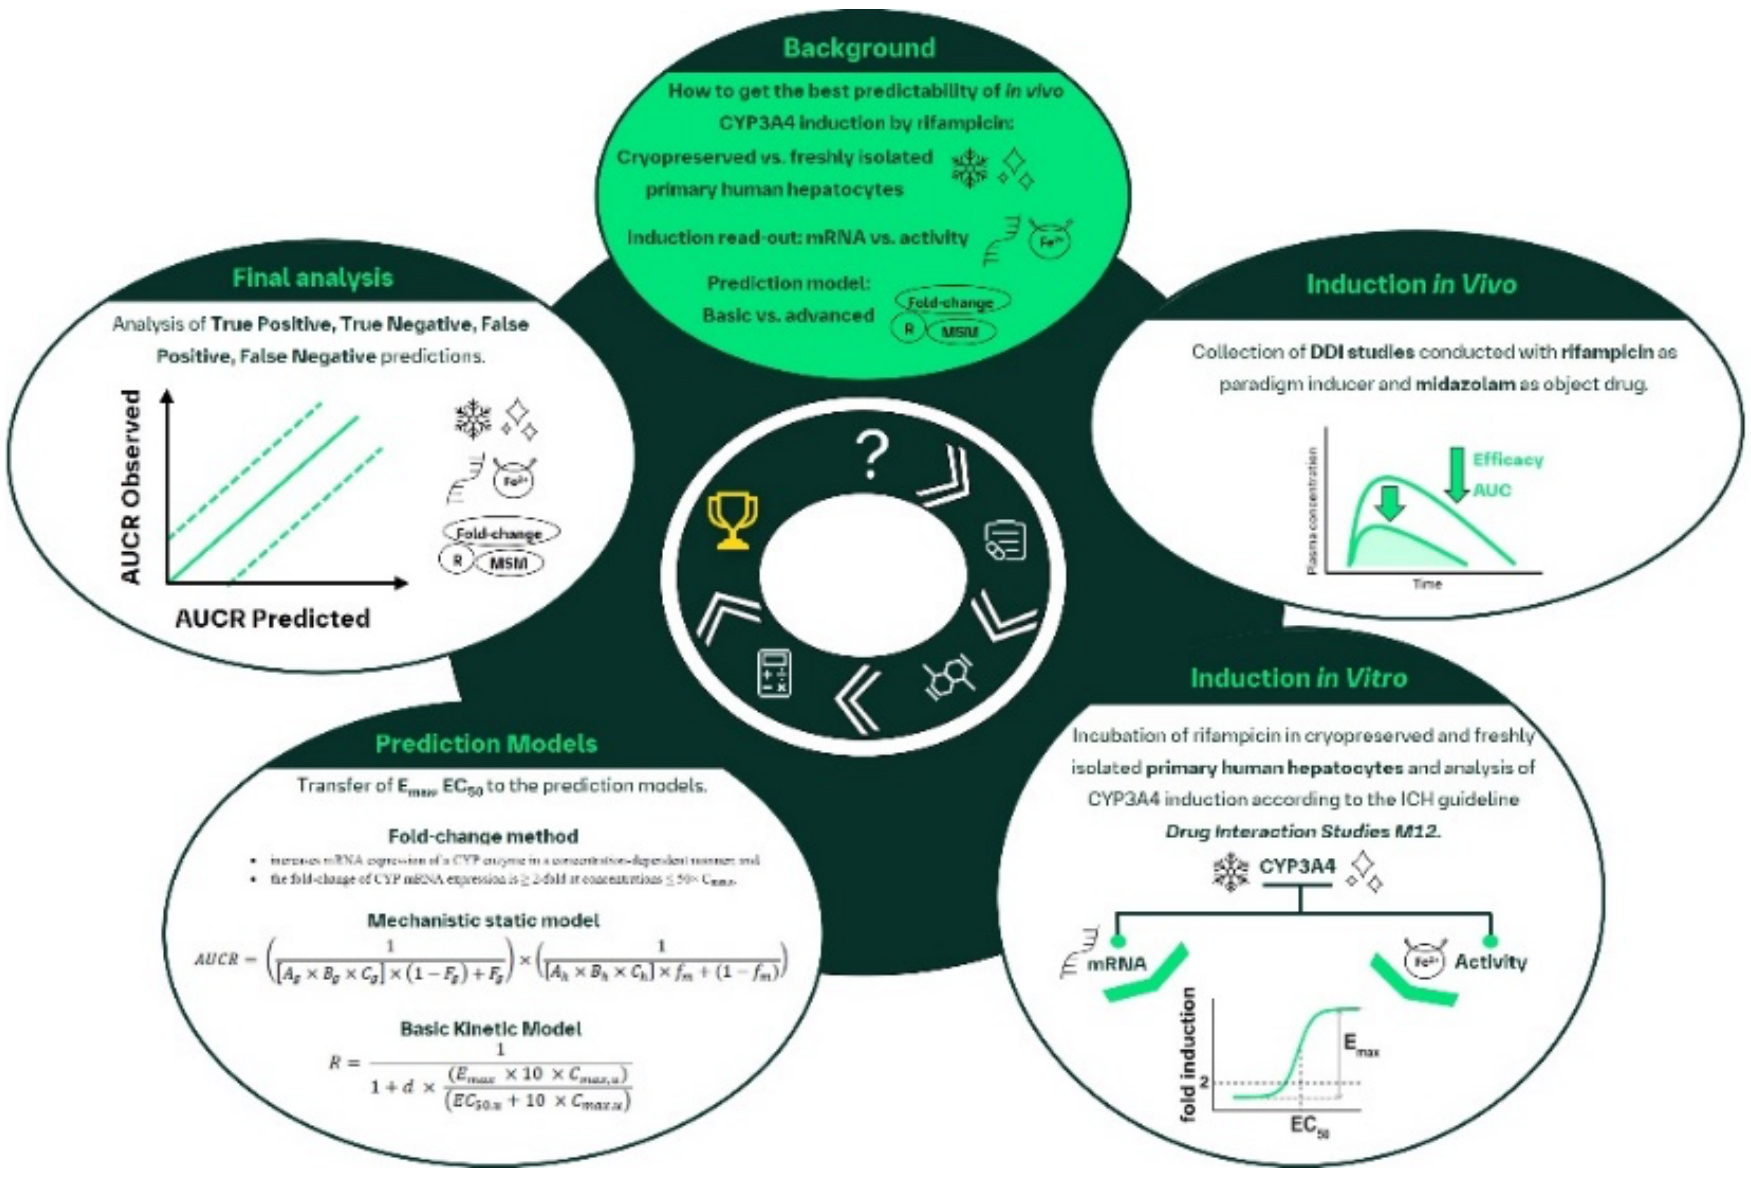
Background
How to get the best predictability of in vivo CYP3A4 induction by rifampicin:
Cryopreserved vs. freshly isolated primary human hepatocytes
Induction read-out: mRNA vs. activity
Prediction model: Basic vs. advanced
Fold-change
R
MSM
Induction in Vivo
Collection of DDI studies conducted with rifampicin as paradigm inducer and midazolam as object drug.
Efficacy
AUC
Plasma concentration
Time
Induction in Vitro
Incubation of rifampicin in cryopreserved and freshly isolated primary human hepatocytes and analysis of CYP3A4 induction according to the ICH guideline Drug Interaction Studies M12.
CYP3A4
mRNA
Activity
fold induction
E_{max}
EC₅₀
Prediction Models
Transfer of E_{max}, EC₅₀ to the prediction models.
Fold-change method
Mechanistic static model
Basic Kinetic Model
Final analysis
Analysis of True Positive, True Negative, False Positive, False Negative predictions.
AUCR Observed
AUCR Predicted
?